Complete regression of solid B16F1melanoma tumors obtained by intratumoral injection of LTX-315 (Oncopore®)

K. CAMILIO[1,2], Ø. REKDAL[1], B. SVEINBJØRNSSON[1,2]
[1]Lytix Biopharma AS, [2]University of Tromsø, Norway

Lytix Biopharma

Introduction

Melanoma is the most serious type of skin cancer, causing about 75% of skin cancer related deaths worldwide. Standard treatment involves surgery, chemo- and radiation therapy, and immunotherapy for metastatic melanoma. However, these treatments have their shortcomings often leading to relapse post treatment.

LTX-315 (Oncopore®) is a chemically modified cationic cytolytic peptide which is equally active against drug-sensitive and drug-resistant cancer cells. Previous *in vivo* studies have demonstrated that treatment of syngeneic murine A20 B-cell lymphomas and CT26WT colon carcinomas with intratumoral (i.t.) injection of LTX-315 resulted in complete tumor regression and long-term protective effect against re-inoculated tumor cells (unpublished data).

B16F1 is a highly aggressive and non-immunogenic murine melanoma syngeneic to the C57BL/6 mouse model.

Aim

The present study was undertaken to investigate whether i.t. injections of LTX-315 induced an antitumor immune response *in vivo* in a highly aggressive murine melanoma tumor model.

LTX-315

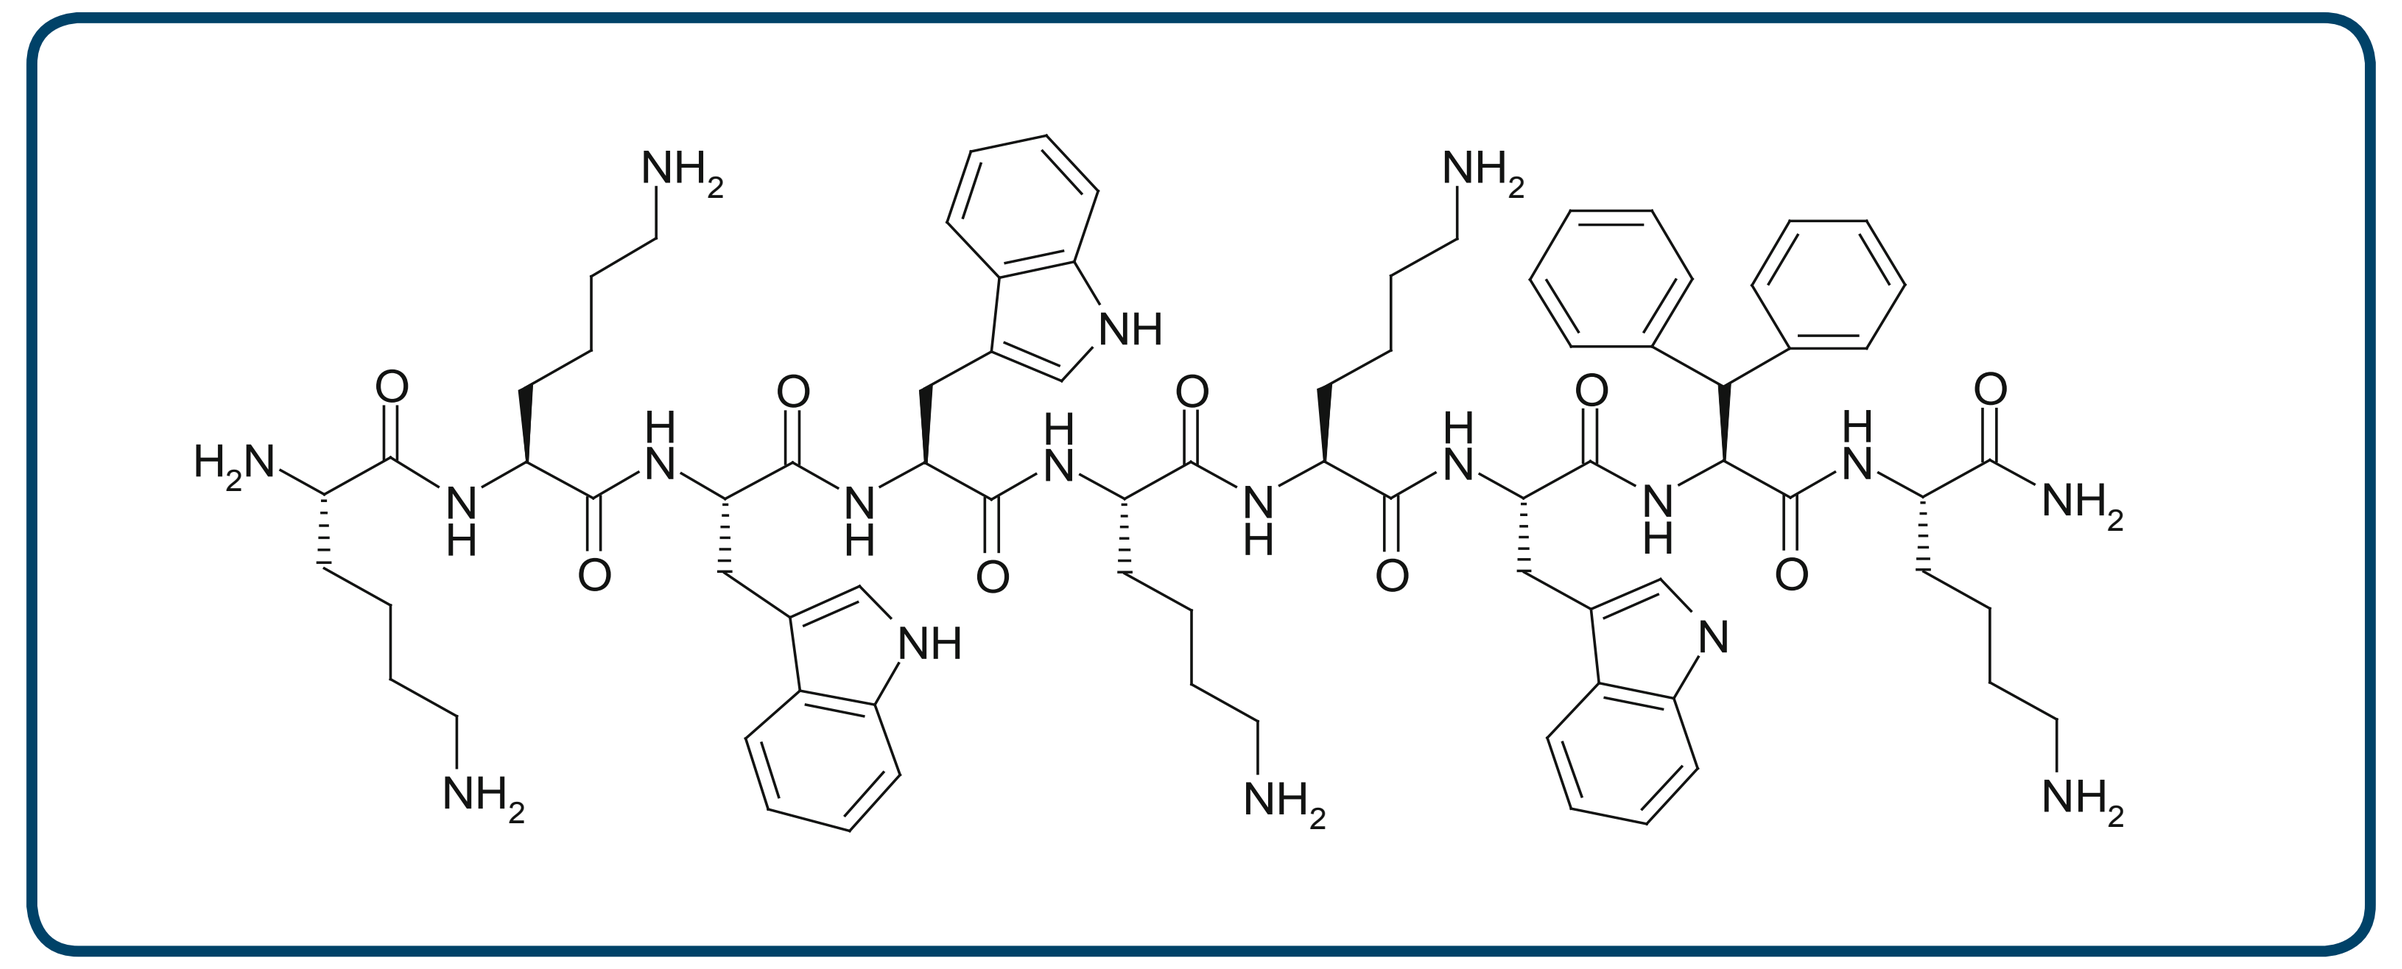

Chemical structure of LTX-315 (Oncopore®)

Methods

In vitro: The colorimetric 3-(4,5-dimethylthiazol-2-yl)-2,5-diphenyltetrazodium bromide (MTT) viability assay was used to investigate the cytotoxic effect of LTX-315 against a selection of human and murine cell lines. Cell lines were incubated with peptide solution for 4 hours.

In vivo: Pre-cultured B16F1 melanoma cells (5 x 104) were subcutaneously inoculated into the abdomen of syngeneic C57BL/6N mice and established tumors treated i.t. with LTX-315 or vehicle (three consecutive injections).

Results

LTX-315 rapidly kills murine and human melanoma cell lines *in vitro.*
MTT cytotoxicity data for LTX-315 against a selection of normal and melanoma cell lines after 4 hours.

Cell line	Organ	Disease	LTX-315 IC_{50} (μM)
MRC-5	Lung	Normal	26
HUV-EC-C	Umbilical vein	Normal	28
RBC	Blood	Normal	>850
B16F1	Skin	Malignant melanoma	20
WM-266-4	Skin	Skin metastasis (primary ephoteloid tumor)	12
A375	Skin	Malignant melonoma	11

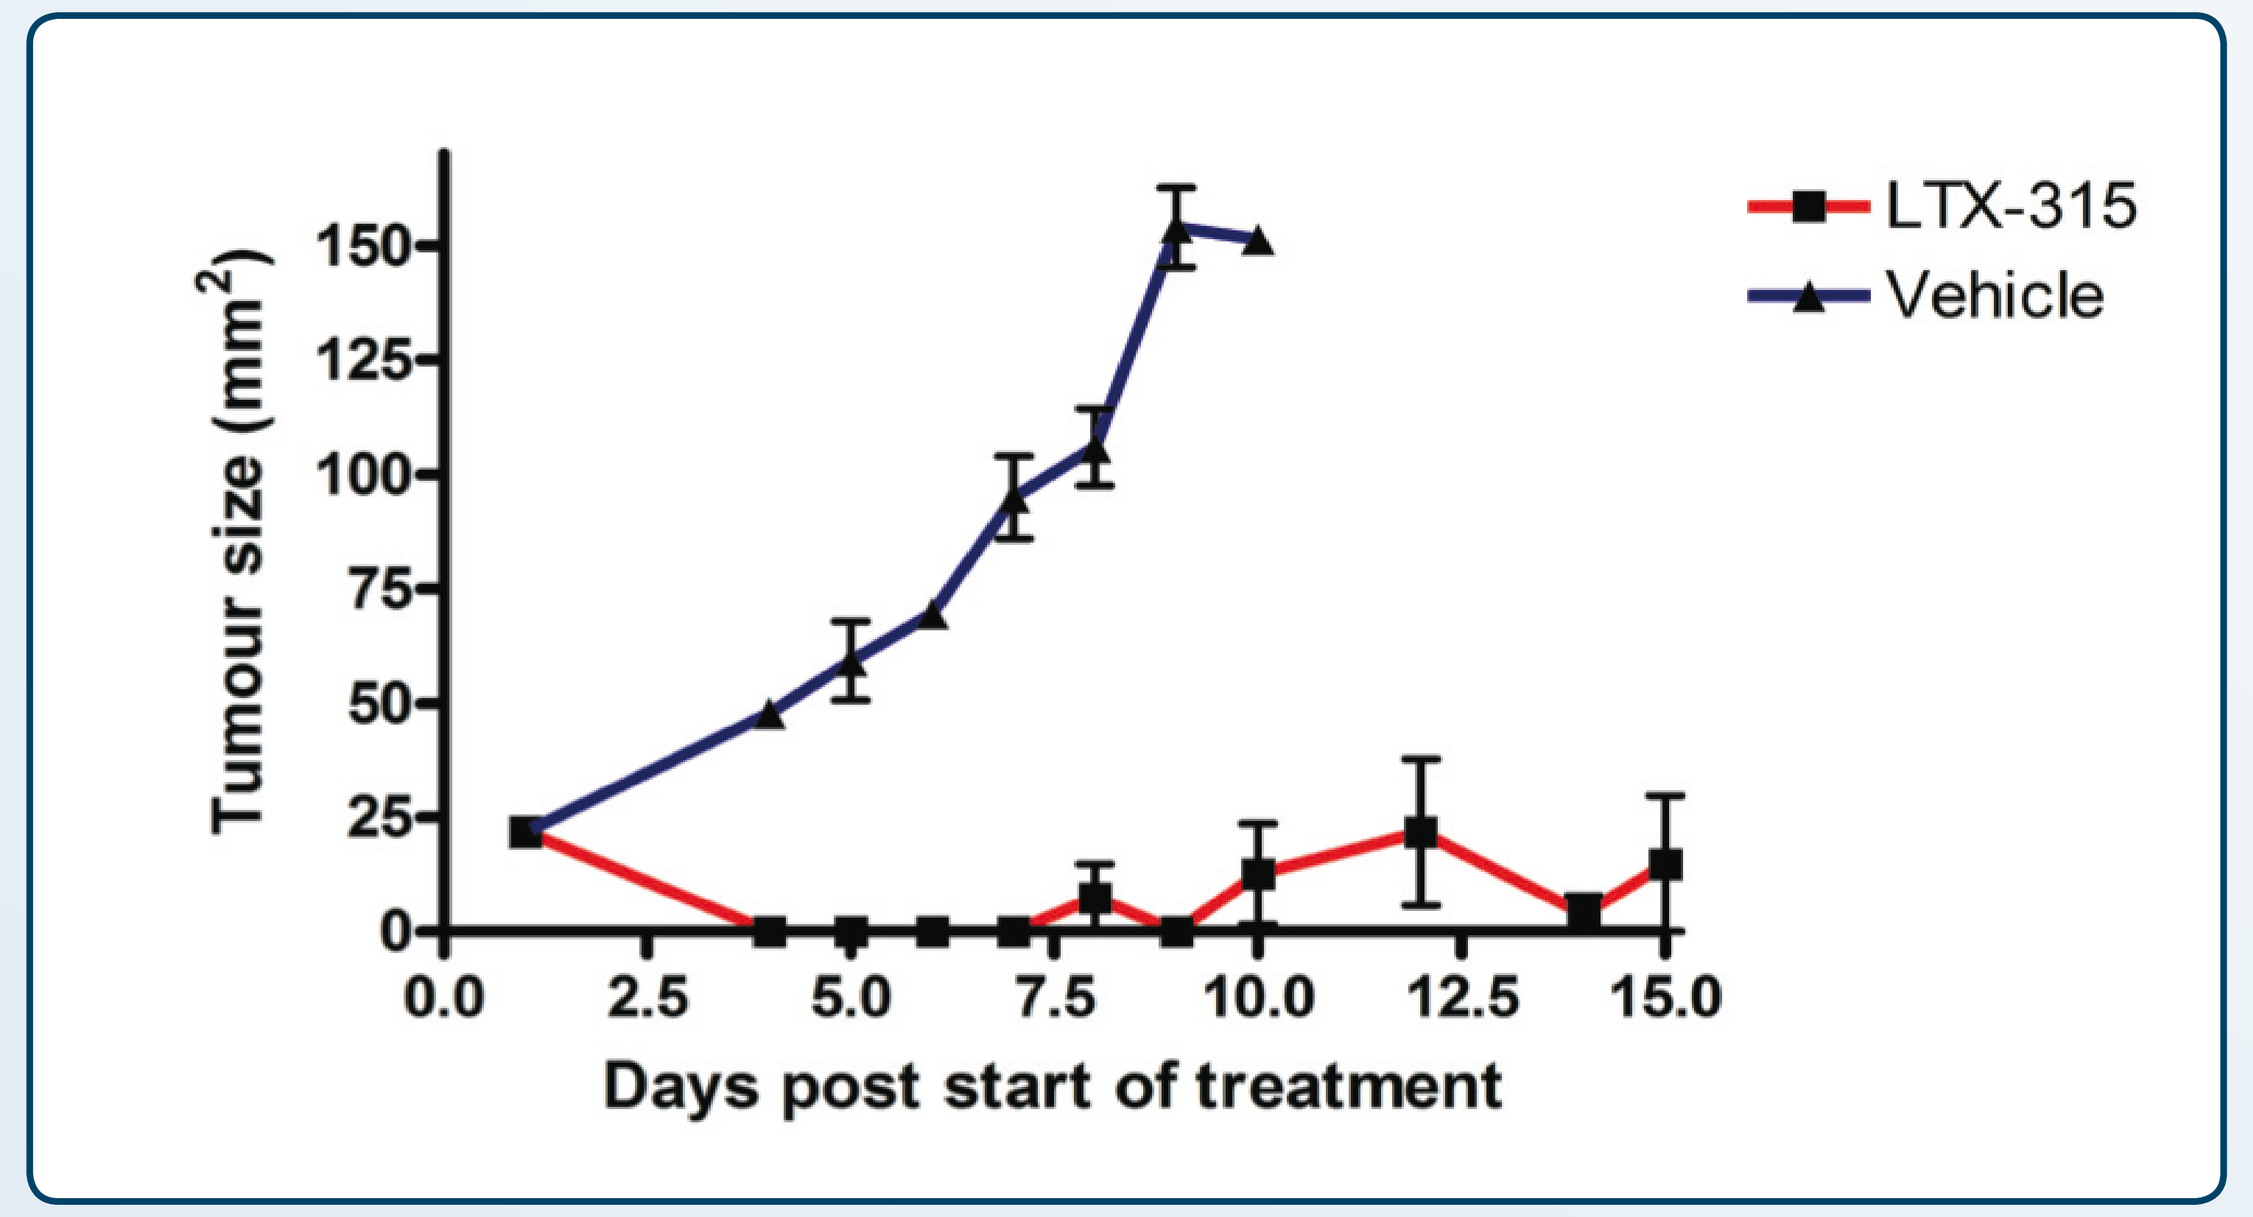
LTX-315
Vehicle
Tumour size (mm²)
150
125
100
75
50
25
0
0.0
2.5
5.0
7.5
10.0
12.5
15.0
Days post start of treatment

LTX-315 eradicates solid subcutaneous B16F1 melanoma tumor *in vivo.*
Growth curves of LTX-315 treated animals (n=8) compared to controls (n=4)

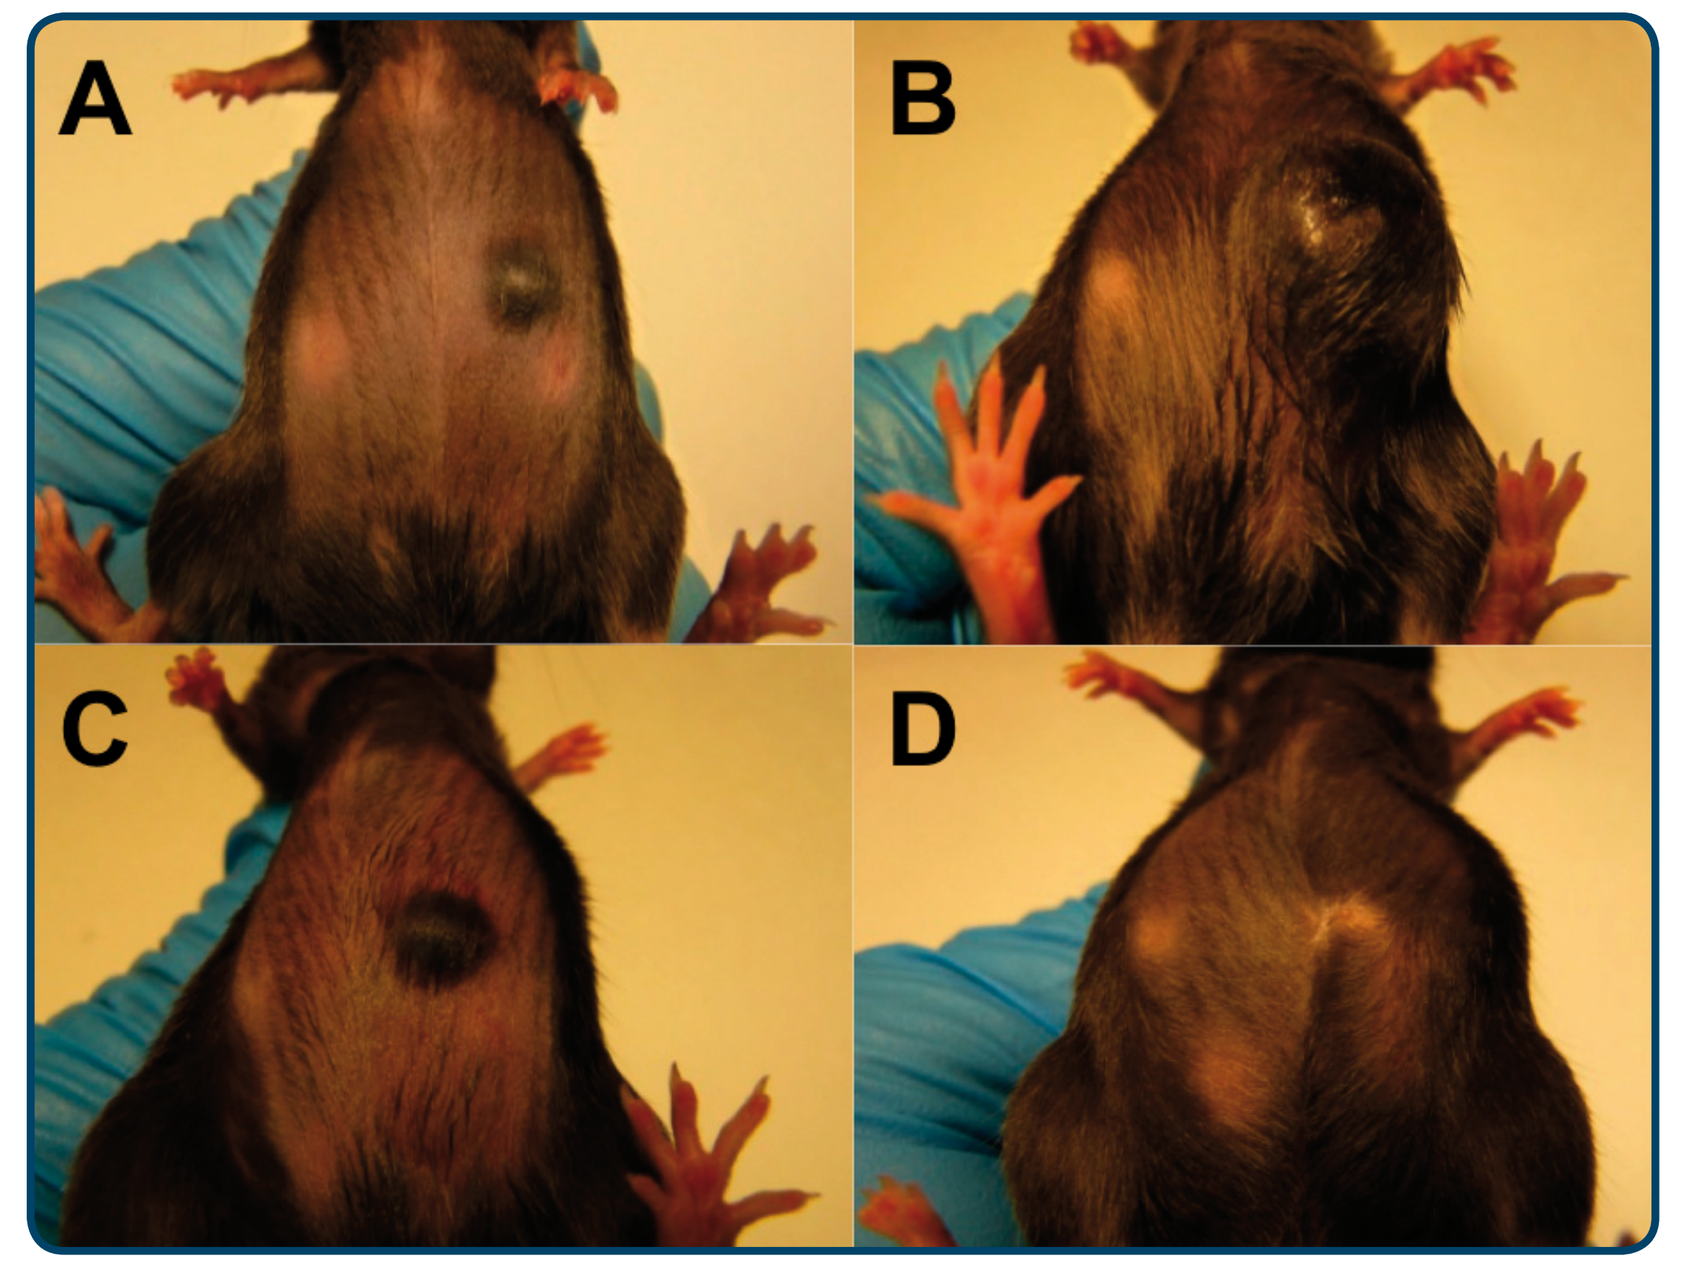
A
B
C
D

Intratumoral treatment with LTX-315 leads to complete regression of solid B16F1 melanoma tumors.
A and B: Animal injected i.t. with vehicle.
C and D: Animal injected i.t. with LTX-315.

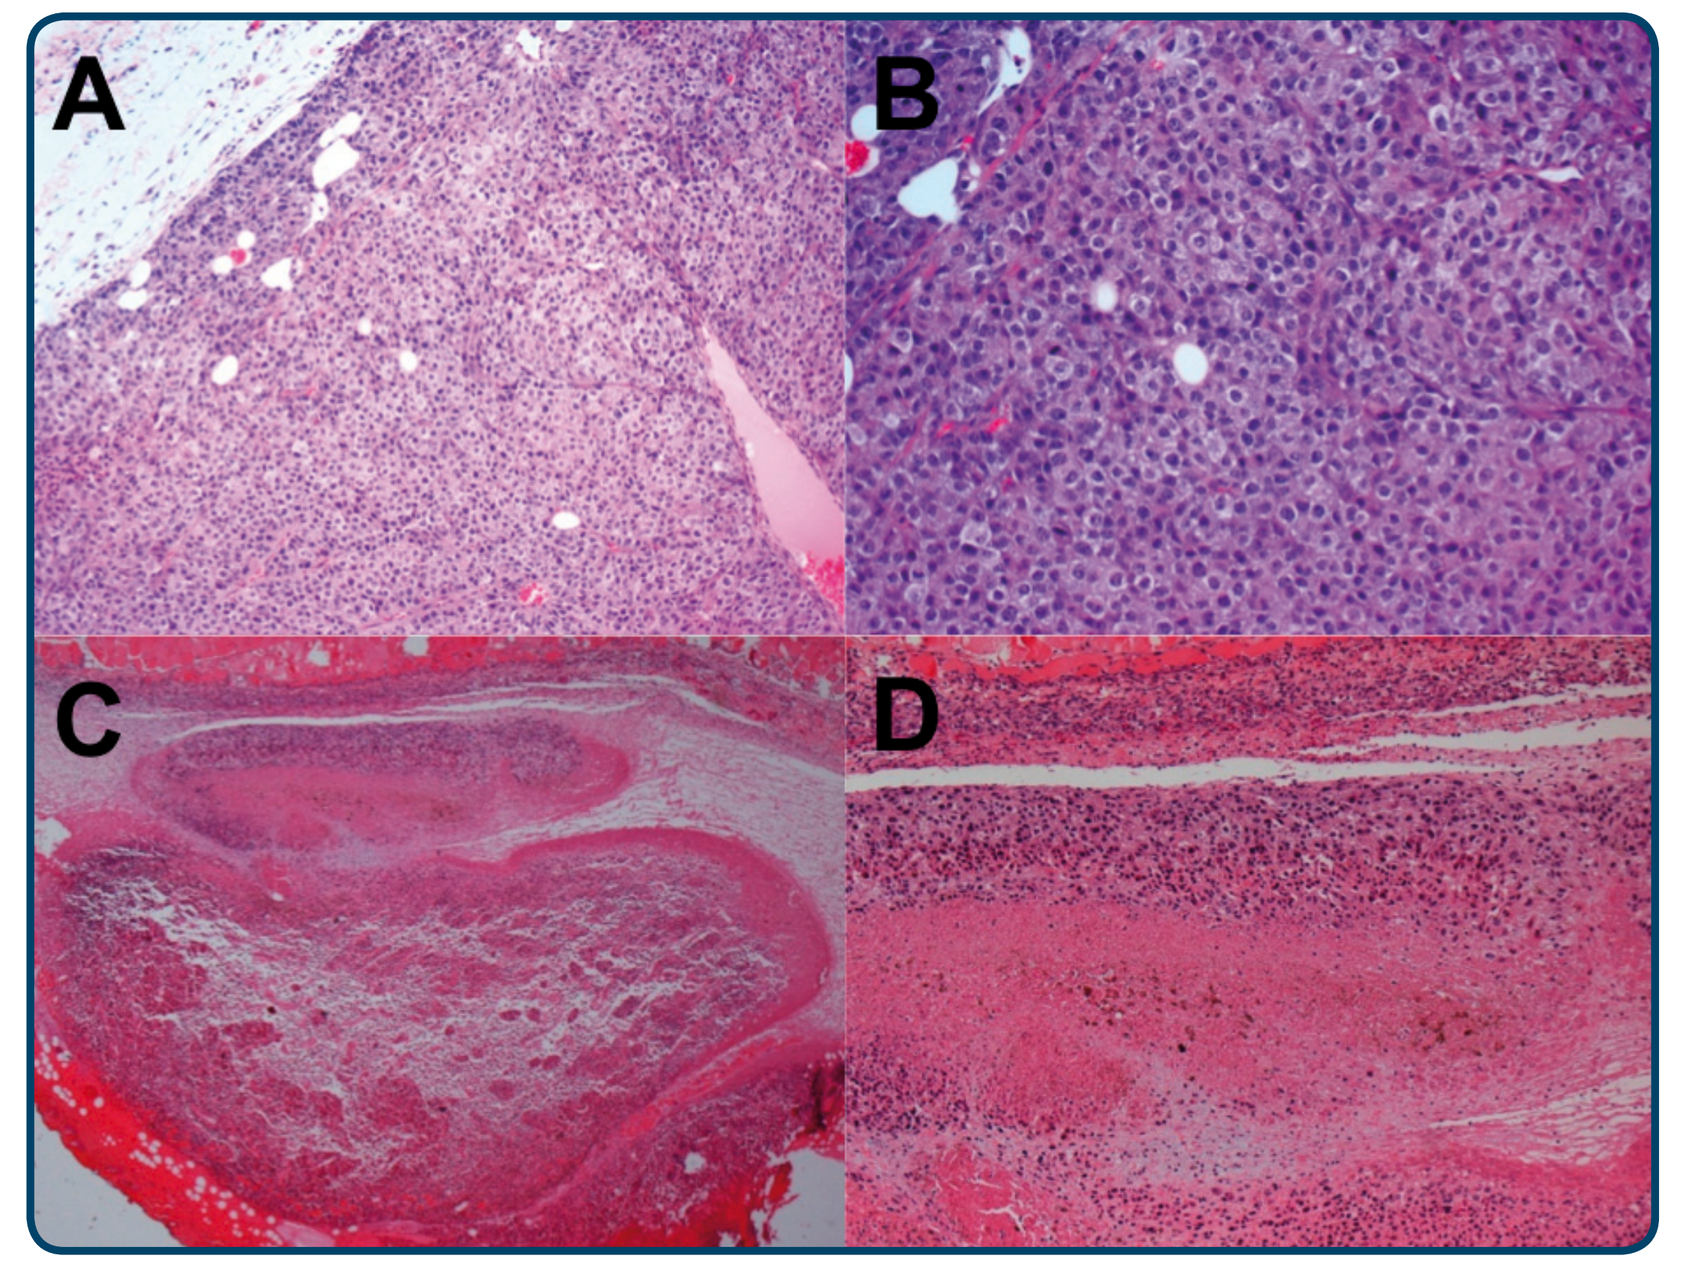
A
B
C
D

Intratumoral treatment with LTX-315 causes massive necrosis and cell infiltration
HE staining of tumor sections from control tumors (A+B) and LTX-315 treated tumors (C+D).

Conclusion

LTX-315 treatment induced complete regression of established B16F1 melanoma tumors.

Intratumoral administration of a cytolytic peptide might represent a novel local treatment for melanoma malignancies.

LTX-315 is currently being investigated in a Phase I clinical study.

This work was supported by:
Norwegian Research Council

Lytix Biopharma AS | P.O. Box 6447 | NO-9294 Tromsø, Norway | E-mail: post@lytixbiopharma.com | Phone: +47 77 67 55 00 | Fax: +47 77 67 55 01